6th MENACTRIMS CONGRESS
December 10-11, 2021
Intercontinental Hotel, Dubai

Hosted by MENACTRIMS
Middle East North Africa Committee for Treatment and Research in Multiple Sclerosis

POSTER PRESENTATION

Economic Analysis for Introduction of Cladribine Tablets as a Treatment for Relapsing Multiple Sclerosis patients with High Disease Activity in Kuwait

Raed Alroughani[1], Samar Farouk Ahmed[2], Amr Abokoura[3], Essam Alsultan[4], Amir Boshra[5], Rawan Alcharif[5], Rita Ojeil[6], Sujata Basu[7]

[1]Department of Medicine, Amiri Hospital, Sharq, Kuwait, Neurology Department, [2]Neurology Department, Ibn Sina Hospital, Kuwait, Neuropsychiatry Department, Faculty of Medicine, Minia University, Egypt, [3]Neurology Division, Mubarak Alkabeer hospital, Kuwait, [4]Head of Pharmacy Department, Ibn Sina hospital, Kuwait [5]Merck Serono Middle East FZ LTD, an affiliate of Merck KgaA, Darmstadt, Germany, [6]HEOR, IQVIA, Middle East, [7]HEOR, IQVIA, India.

INTRODUCTION

- Multiple sclerosis (MS) is a chronic demyelinating neurological disorder characterized by loss of sensory and motor functions.[1]
- In 2019, the estimated prevalence and incidence of MS in Kuwait per 100,000 people was 104.9 and 5.39, respectively.[2]
- Cladribine tablets is the first oral short course treatment approved for the highly active relapsing multiple sclerosis across various geographies.[3]

OBJECTIVES

To estimate the budget impact (BI) of introducing Cladribine Tablets compared to other disease-modifying drugs for the treatment of High Disease Active (HDA) relapsing MS (RMS) patients from payer's perspective in Kuwait

METHODS

• This BI analysis was performed using an excel-based BI model. Three scenarios, 'MS treatments without cladribine' and 'MS treatments with cladribine gradual introduction' and 'MS treatments with full replacement with cladribine' were assessed over a five-year time horizon based on the perspective of MS experts in Kuwait. The comparators included dimethyl fumarate, fingolimod, natalizumab, ocrelizumab and alemtuzumab.

• The model inputs included total number of HDA-RMS patients, market shares, and costs associated with drug administration, monitoring, adverse events management, and relapse therapy. The inputs were retrieved from literature and/or obtained by interviewing the key experts in Kuwait representing governmental sectors. All costs were presented in Kuwaiti Dinars (KWD).

• The results of the model were presented to the interviewed MS experts and payers for validation. The outcome of the BI model are endorsed by the authors.

Figure 1. *Detailed Methodology*

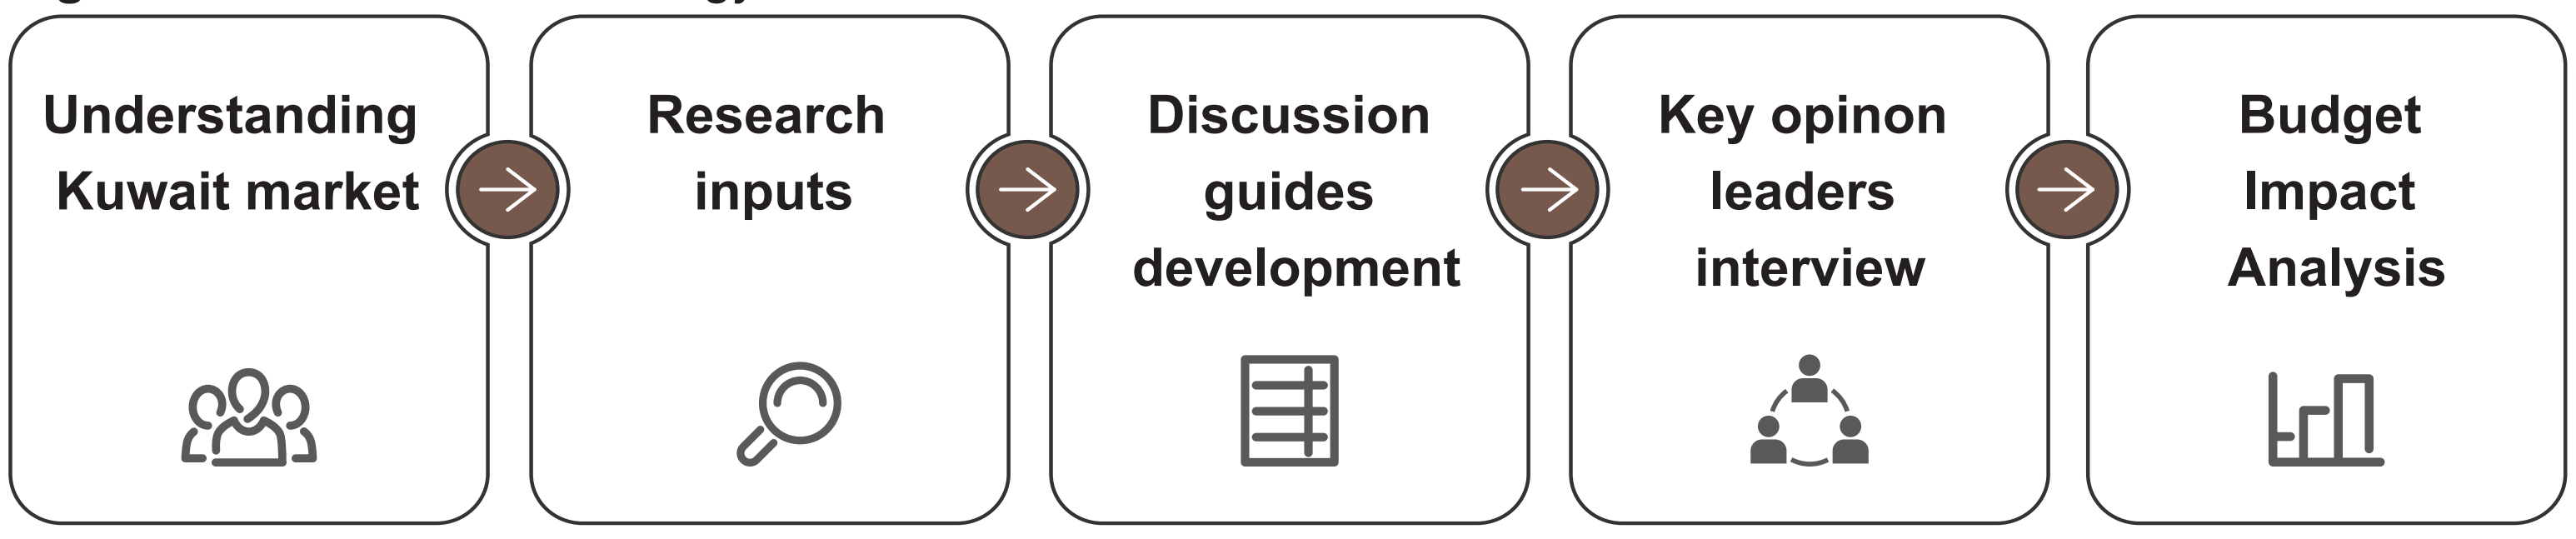
Understanding Kuwait market
Research inputs
Discussion guides development
Key opinon leaders interview
Budget Impact Analysis

RESULTS

- A gradual introduction of cladribine tablets for the treatment of HDA-RMS (starting by 5% in first year and reaching up to 18% market share in year five) resulted in an estimated cost savings of 4.8% (KWD 984,282). Whereas with a 100% replacement of existing treatments with cladribine tablets, the estimated cost savings was 28.5% (KWD 5,9 millions). ***(Figure 2)***

- The major contributors for cost savings in HDA budget were drug acquisition cost, expenditure associated with adverse event and rescue treatment. ***(Figure 3)***

- The cumulative treatment cost per HDA- RMS patient (including all cost inputs) over 4 years with cladribine tablets was KWD 21,268 which is the least option compared most of the currently used disease modifying drugs (DMDs). ***(Figure 4a)***

- On comparison with cladribine tablets, the incremental difference in per patient treatment cost for HDA patient over 4 years, was highest for fingolimod by 92%, natalizumab by 81% then ocrelizumab and alemtuzumab by 58%. ***(Figure 4b)***

Figure 2. *Cumulative economic impact on HDA-RMS population over 5 years (KWD)*

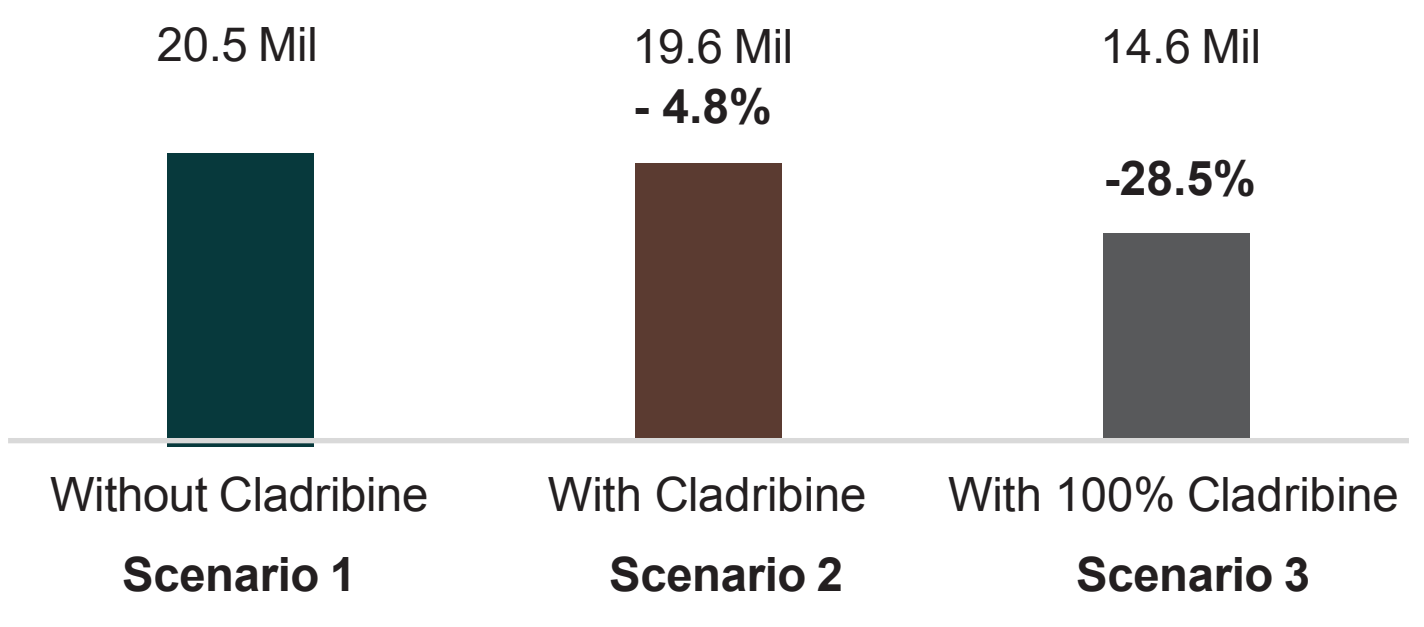
20.5 Mil
19.6 Mil
- 4.8%
14.6 Mil
-28.5%
Without Cladribine
Scenario 1
With Cladribine
Scenario 2
With 100% Cladribine
Scenario 3

Figure 3. *Contribution of individual cost inputs on HDA population (in Scenario 2)*

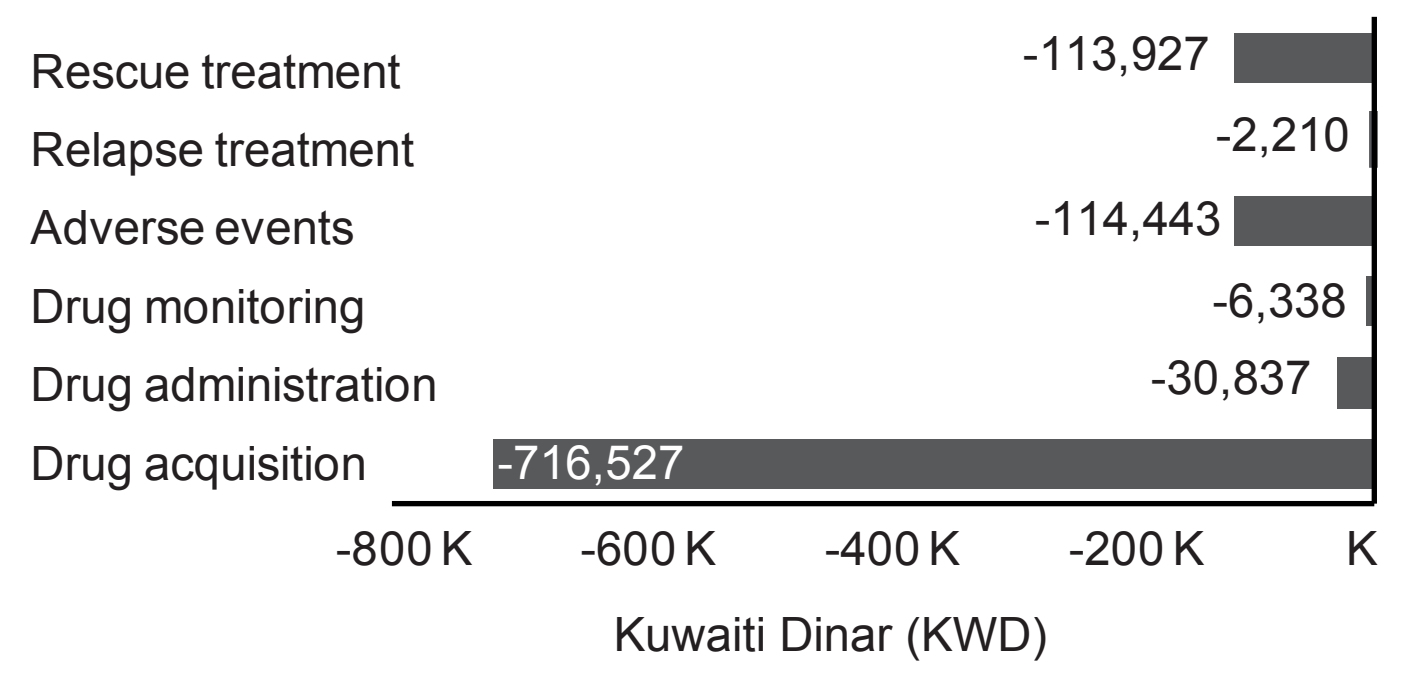
Rescue treatment
-113,927
Relapse treatment
-2,210
Adverse events
-114,443
Drug monitoring
-6,338
Drug administration
-30,837
Drug acquisition
-716,527
-800 K
-600 K
-400 K
-200 K
K
Kuwaiti Dinar (KWD)

Figure 4a. *Cumulative direct and indirect cost inputs of Cladribine tablets for an individual HDA-RMS patient over 4 years (KWD)*

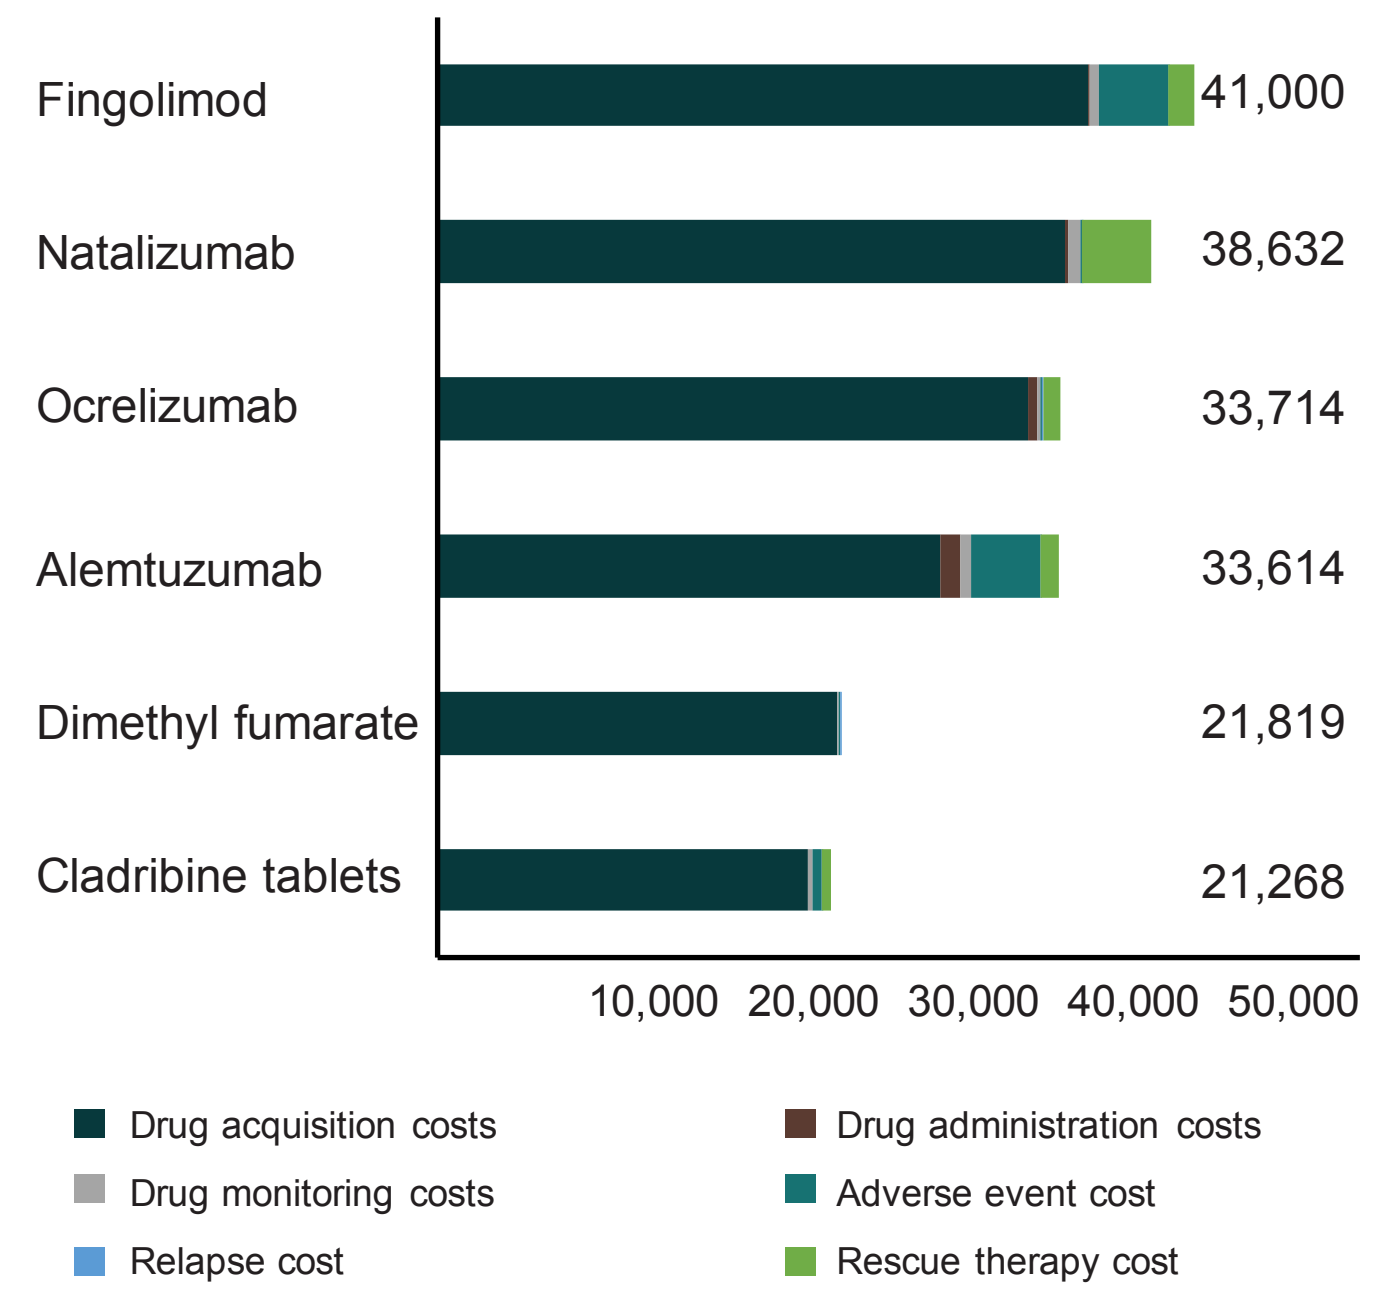
Fingolimod
41,000
Natalizumab
38,632
Ocrelizumab
33,714
Alemtuzumab
33,614
Dimethyl fumarate
21,819
Cladribine tablets
21,268
10,000
20,000
30,000
40,000
50,000
Drug acquisition costs
Drug administration costs
Drug monitoring costs
Adverse event cost
Relapse cost
Rescue therapy cost

Figure 4b. *Incremental Difference in per patient treatment cost between Cladribine tablets and DMDs over 4 years (KWD)*

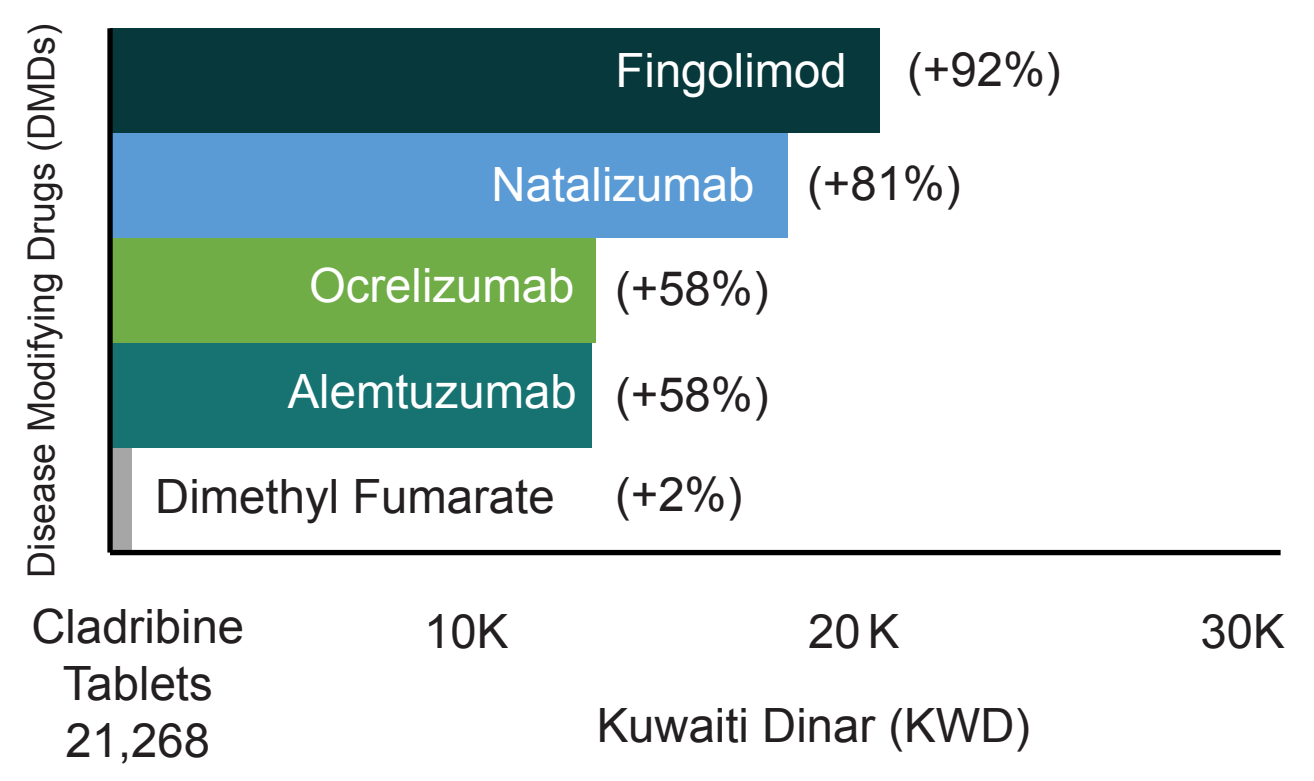
Disease Modifying Drugs (DMDs)
Fingolimod
(+92%)
Natalizumab
(+81%)
Ocrelizumab
(+58%)
Alemtuzumab
(+58%)
Dimethyl Fumarate
(+2%)
Cladribine Tablets 21,268
10K
20 K
30K
Kuwaiti Dinar (KWD)

CONCLUSION

Introduction of cladribine tablets in Kuwait as a treatment option in HDA relapsing MS patients shows cost-saving from the payer's perspective. The savings in budget improved with an increase in the cladribine tablets market share

References: 1. Number of people with MS | Atlas of MS [Internet]. [cited 2021 Apr 22]. Available from: https://www.atlasofms.org/map/global/epidemiology/number-of-people-with-ms; 2. Alroughani R et al. The prevalence of multiple sclerosis continues to increase in Kuwait. Mult Scler Relat Disord. 2019 Jul;32:74–6; 3. Giovannoni G, et al. Safety and efficacy of Cladribine tablets in patients with relapsing–remitting multiple sclerosis: Results from the randomized extension trial of the CLARITY study. Mult Scler J. 2018 Oct;24(12):1594–604

Acknowledgement: The authors would like to thank all contributors for their commitment and dedication to the goals of MS treatment. The medical writing support was provided by Steffy Martin Luther King and Smitha Sreedharan (IQVIA, India) . The authors are fully responsible for all content and editorial decisions, were involved at all stages of poster development, and have approved the final version

Disclosure: This study was conducted by IQVIA Middle East and Africa. Funding was provided by Merck Serono Middle East FZ LTD, an affiliate of Merck KGaA (CrossRef Funder ID: 10.13039/100009945)